Physicochemical Stability of Anhydrous Aspirin Oral Pediatric Suspensions: Comparison of Two Mixing Methods

Courtaney Davis, Kendice Ip, Matt Martin, Melissa Merrell Rhoads, Fabiana Banov and Maria Carvalho

PCCA, 9901 South Wilcrest Drive, Houston, TX 77099, USA

Poster ID #2538

Introduction

Aspirin is an antiplatelet drug with limited use in pediatrics because it has been associated with Reye's syndrome. It is therefore not licensed for use in children under 16 years, but it may be specifically indicated off-label in conditions such as: Kawasaki disease, prophylaxis of clot formation after cardiac surgery, and prophylaxis of stroke in children at high risk. Dosing is weight-dependent and requires ongoing adjustments throughout treatment [1]. Therefore, a versatile oral liquid dosage form is essential to meet the evolving needs of pediatric patients. The purpose of this study is to investigate the physicochemical stability of anhydrous aspirin oral pediatric suspensions (10 mg/mL and 20 mg/mL), prepared using two different mixing methods and stored at both room temperature and refrigerated conditions.

Methodology

Oral pediatric suspensions for aspirin 10 mg/mL and 20 mg/mL were prepared by mixing the aspirin powder with the same amount of glycerin (mL), and adding the mixture to Anhydrous SuspendIt®, a proprietary oral vehicle – USP (United States Pharmacopoeia) verified excipient - with water activity less than 0.6 (anhydrous) which exhibits self-emulsifying properties and thixotropic effects [2]. The oral suspensions were split into two batches (100 mL and 450 mL) and were further mixed using two different methods: Electronic Mortar & Pestle (EMP) (Unguator® e/s) (Figure 1), which is typically used for small-batch extemporaneous preparations, and the IKA® stirrer (Figure 2) which is suited for larger-scale compounding, commonly in hospital pharmacy settings. The resulting oral pediatric suspensions were evenly distributed into prescription oval amber plastic bottles and stored at controlled room temperature (25°C) and refrigerated (5°C) conditions for 6 months. At 12 pre-determined time points, a bottle from each strength and mixing method was taken from the storage conditions and shaken vigorously. A study sample from each was then withdrawn for physicochemical stability testing. Samplings were done at top, middle, and bottom of the bottle at the time of testing. Physical characterization consisted of observing all samples for appearance and color. pH testing was not performed, as the suspensions are anhydrous. Viscosity and sedimentation were not tested in this study due to the large amount of sampling required. The chemical characterization consisted of assay testing employing a stability-indicating High Performance Liquid Chromatography (HPLC) method developed and validated by Eagle Analytical Laboratories (Texas, US). The method was validated for the assay determination of aspirin in the anhydrous suspensions. Identification and quantitation of the impurities, degradants, and excipients were not part of the purpose of the method validation.

Results & Discussion

The aspirin oral pediatric suspensions exhibited a uniform pale-yellow color which remained stable throughout the study. The chromatographic assay method demonstrated to be linear, precise, accurate, robust and suitable, as well as stability-indicating. The validation of the method has demonstrated the specificity and selectivity of the aspirin peak being well separated from all other impurity, degradant, and excipient peaks. The potency of the suspensions was similar, which shows that both compounding methods are suitable and may equally ensure quality preparations. Samples stored at room temperature exhibited instability early in the study and thus it is recommended to refrigerate the aspirin oral pediatric suspensions for extended stability (Figure 3). Under refrigerated conditions, the higher strength samples presented better stability than the lower strength samples (Table 1). A higher excipient-to-drug ratio in low-strength formulations may increase the susceptibility of the drug to degradation, among other possible reasons.

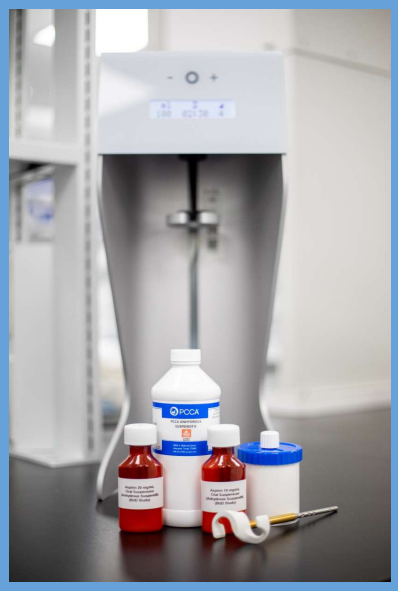

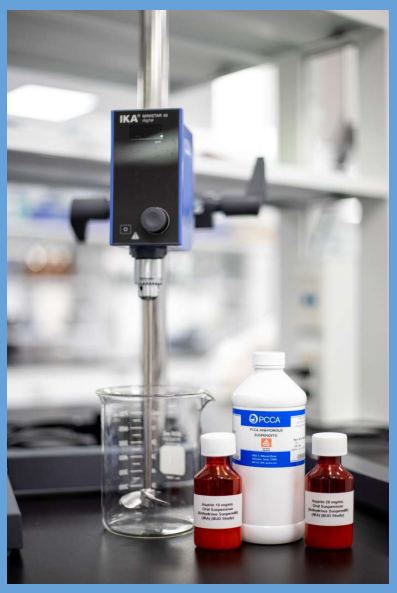

Figure 1 (left). *Electronic Mortar & Pestle (EMP): Unguator® e/s.*
Figure 2 (right). *IKA® stirrer.*

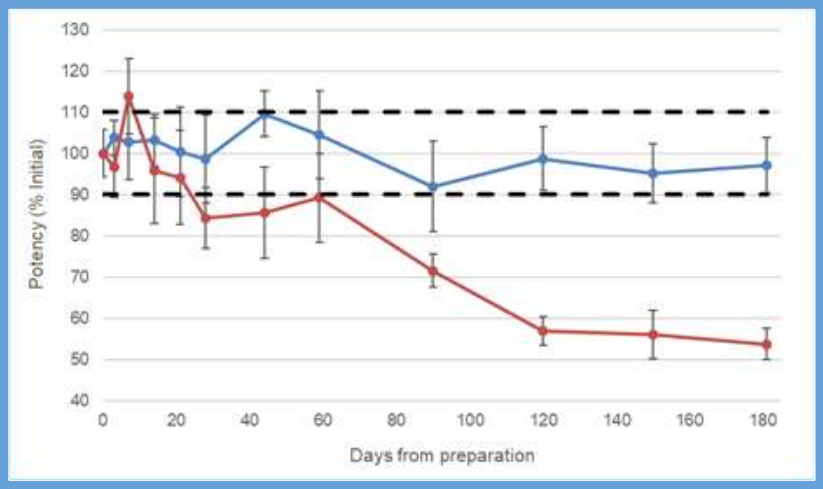
130
120
110
100
90
80
70
60
50
40
0
20
40
60
80
100
120
140
160
180
Potency (% Initial)
Days from preparation

Figure 3. *Potency over time for the aspirin 20 mg/mL oral pediatric suspensions prepared using the EMP (Unguator® e/s). Blue and red lines represent refrigerated and controlled room temperature, respectively. Dashed lines represent the upper and lower limits.*

Table 1. *BUD for the aspirin 10 mg/mL and 20 mg/mL oral pediatric suspensions prepared using the two mixing methods, and stored at both refrigerated and controlled room temperature.*

Mixing Methods	Aspirin 10 mg/mL		Aspirin 20 mg/mL	
	T=5°C	*T=25°C*	*T=5°C*	*T=25°C*
EMP (Unguator® e/s)	90 days	3 days	180 days	21 days
IKA® stirrer	88 days	7 days	180 days	14 days

Conclusion

The beyond-use-date (BUD) of the anhydrous aspirin oral pediatric suspensions stored at refrigerated conditions is: 90 days (EMP) and 88 days (IKA) for the lower strength (10 mg/mL) and 180 days (both methods) for the higher strength (20 mg/mL).

1. Paediatric Formulary Committee (2022) British National Formulary for Children 2022–2023 [online]. London: BMJ Publishing Group and Pharmaceutical Press. Available at: https://bnfc.nice.org.uk/ (Accessed: 20 June 2025).
2. Banov, D.; Liu, Y.; Ip, K.; Shan, A.; Vu, C.; Zdoryk, O.; Bassani, A.S.; Carvalho, M. Analysis of the Physical Characteristics of an Anhydrous Vehicle for Compounded Pediatric Oral Liquids. Pharmaceutics 2023, 15, 2642. https://doi.org/10.3390/pharmaceutics15112642.